Version 3.0

MULTIPLE MYELOMA IN A SWISS POPULATION

REAL-WORLD DRUG REGIMENS AND COSTS OF TREATMENT (2012 - 2017)

Zurich University of Applied Sciences

Eichler K [1], Rapold R [2], Wieser S [1], Gratwohl A [3], Reich O [2], Blozik E [2]

[1] Winterthur Institute of Health Economics, Zurich University of Applied Sciences, Winterthur, Switzerland
[2] Helsana Insurances, Zurich, Switzerland;
[3] Medical Faculty University Hospital of Basle, Switzerland

corresponding address: eich@zhaw.ch

Background Novel drugs are dynamically changing treatment for multiple myeloma (MM). They have improved prognosis in clinical studies but are expensive. Little is known about up-to-date real-world drug application, costs and death rates.

Research question

What are current multiple myeloma (MM) treatment regimens, associated costs and mortality in a Swiss real-world setting of MM patients from 2012 to 2017?

Methods

Design: Retrospective observational cohort analysis (2012-2017)

Data: Claims database of a major Swiss health insurance company (population coverage: 14%).

Inclusion criteria: Patients with MM, defined by ICD-10 diagnoses (primary criterion), and prescribed MM-specific drugs (secondary criterion).

Analysis: We defined a hierarchy of drug regimens:

1) Proteasome inhibitor (PI)-based (e.g. bortezomib)

2) IMID-based (e.g. lenalidomide)

3) Chemotherapy (CHEMO)-based (e.g. bendamustin)

4) Monoclonal antibody (MAB)-based (e.g. daratunumab)

Based on this hierarchy, we analysed real-world treatment patterns of incident patients from 2015 to 2017*. Direct medical costs of mandatory health insurance were analysed in 2017 Swiss Francs (CHF; third party payer perspective).

*For example, if bortezomib and lenalidomide were applied within one drug regimen, the regimen was defined as PI-based regimen; if lenalidomide and bendamustin were applied within one drug regimen, the regimen was defined as IMID-based regimen.

Key Results

Patients: We identified n=1054 prevalent MM patients (2012-2017) and n=378 incident MM patients (2015-2017; 47% men; age >75yr.: 51%). The annual number of prevalent patients increased from n=314 to n=645.

Drug patterns: PI-based regimens were the most frequent first line approach (76.0%), followed by IMID-based (21.9%) and CHEMO-based (2.1%; **Figure 1**).

Stem cell transplantation:15.3% of prevalent patients were treated with autologous hematopoietic stem cell transplantation.

Costs: Costs per treatment line varied between CHF 81’352 (PI-based) and CHF 73’495 (IMID-based).

- Mean daily costs under MM treatment increased stepwise from 2012 (CHF 209) to 2017 (CHF 254): relative increase of 21.5%.
- Extrapolation of annual direct medical costs in Switzerland for seven novel MM drugs: CHF 60.1 Mio in 2012 and CHF 118.6 Mio in 2017 (relative increase: 97.3%).
- MM drug costs accounted for 45% of total annual costs of myeloma patients (**Figure 3**).

Mortality: Annual death rates decreased from 18.6% to 15.5% (risk ratio: 0.83; 95%-CI: 0.63 to 1.10; p for trend: 0.03; **Figure 3**).

Strengths and Limitations

(1) Real-world data of unselected MM patients
(2) No clinical charts were available (no info about co-morbidities)
(3) Some data my be missing for patients who entered the study late (e.g. in 2017).

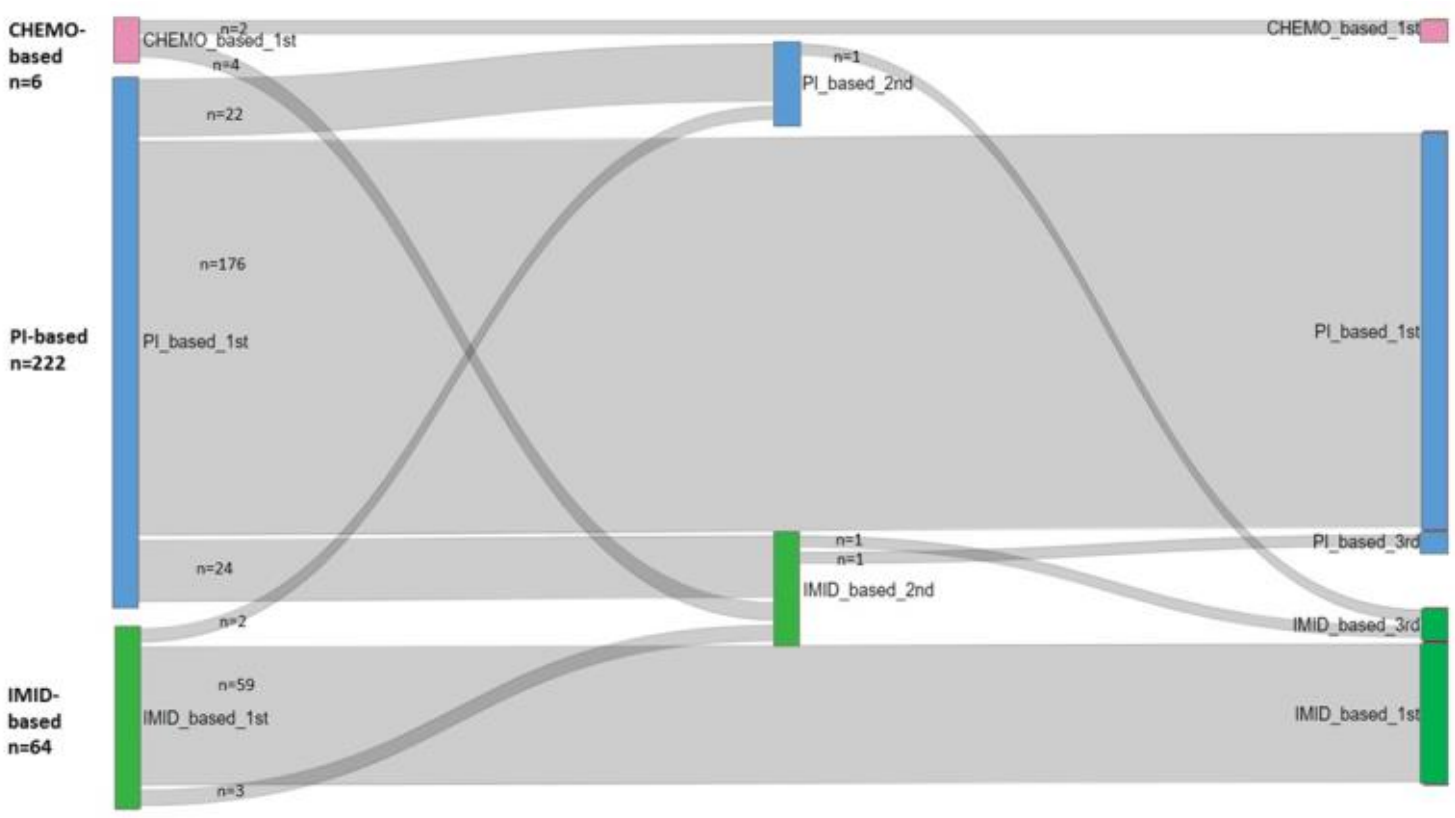
CHEMO-based n=6
PI-based n=222
IMID-based n=64
CHEMO_based_1st
PI_based_1st
IMID_based_1st
PI_based_2nd
IMID_based_2nd
PI_based_3rd
IMID_based_3rd
n=2
n=4
n=22
n=176
n=24
n=2
n=59
n=3
n=1
n=1
n=1

Figure 1: MM drug treatment patterns. Analysis based on 292 of 378 incident MM patients from 2015 to 2017 (mean follow-up 2.1 yr.). Sequence of treatment lines according to 1st-line drug regimen groups (PI-based: mean duration of 1st line: 218 days; IMID-based: 214 days; CHEMO-based: 64 days). 237 of 292 patients (81.2%) remained in their 1st-line treatment. For 55 patients (18.8%) a 2nd-line treatment was started (3rd line: 3 of 292 patients).
Only four patients were treated with MAB-drugs, three times as part of PI-based regimens and once as part of an IMID-based regimen.

Figure 2: Pattern of annual costs of MM patients. Outpatient MM drug costs (CHF 26’224) accounted for about 45% of total annual costs of all-diagnoses inpatient and outpatient services of myeloma patients (CHF 58’401).

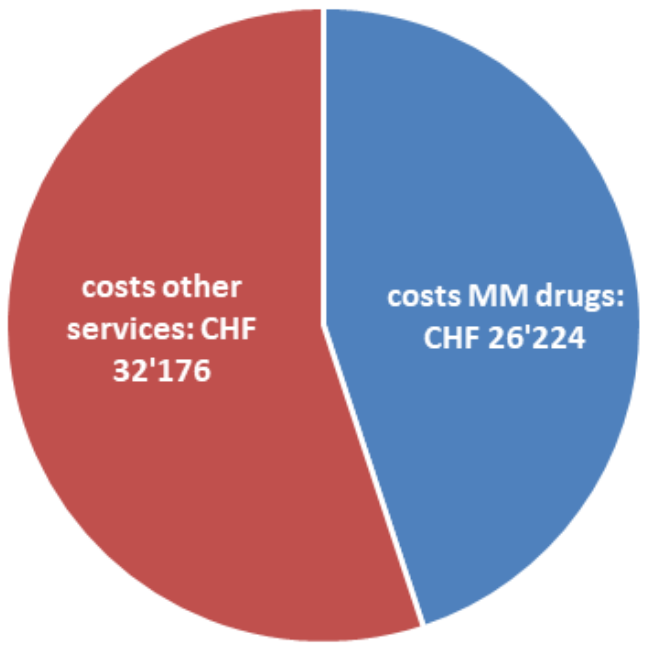
costs other services: CHF 32'176
costs MM drugs: CHF 26'224

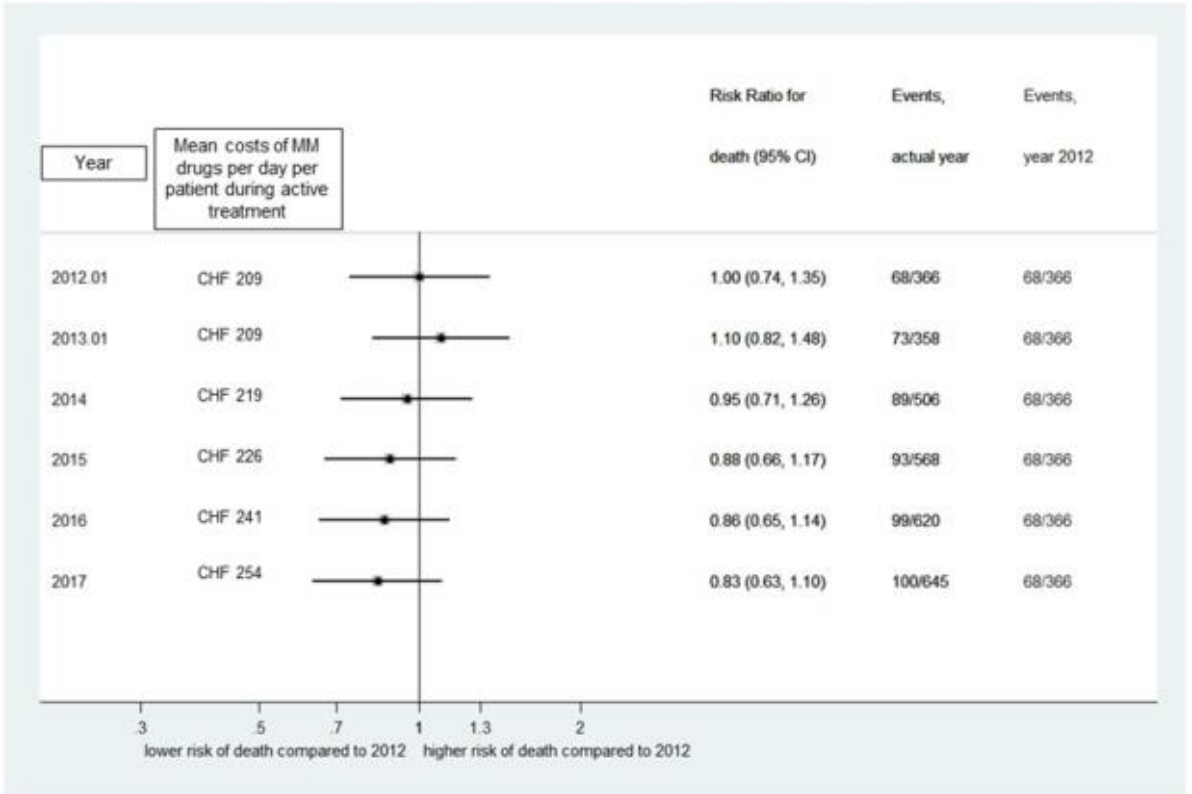
Year
Mean costs of MM drugs per day per patient during active treatment
Risk Ratio for death (95% CI)
Events, actual year
Events, year 2012
2012.01 CHF 209 1.00 (0.74, 1.35) 68/366 68/366
2013.01 CHF 209 1.10 (0.82, 1.48) 73/358 68/366
2014 CHF 219 0.95 (0.71, 1.26) 89/506 68/366
2015 CHF 226 0.88 (0.66, 1.17) 93/568 68/366
2016 CHF 241 0.86 (0.65, 1.14) 99/620 68/366
2017 CHF 254 0.83 (0.63, 1.10) 100/645 68/366
.3 .5 .7 1 1.3 2
lower risk of death compared to 2012
higher risk of death compared to 2012

Figure 3: Risk ratio for death and daily treatment costs under MM treatment compared to 2012. Analysis based on 3061 observed patient years of prevalent MM patients from 2012 to 2017. All risk ratios are calculated for reference year 2012.

Conclusions An increasing prevalent population of MM patients in combination with increasing costs per day under treatment lead to a growing budget impact for the Swiss social insurance system.
Our data call for an introduction of “Coverage with Evidence Development” (CED) reimbursement. This form of reimbursement provides continued access to novel MM drugs in Switzerland and might trigger the establishment of registries and foster further research.

Competing interests: The authors KE, RR, SW, EB, OR declare that they have no competing interests.
Funding: The study was in part financially supported by Amgen Switzerland AG, Zug, Switzerland. The funding body commented on the final draft of the manuscript but did not make final decisions regarding the design of the study, the data collection, the analysis and the interpretation of results.